Pharmacokinetic and Pharmacodynamic Profiles of Orally, Duodenally and Subcutaneously Delivered Insulin in Beagle Canines

[1]Ya-zhuo Li, [1]Wei-dang Wu, [1]Rong Zeng, [2]Yael Greenberg-Shushlav, [3]Miriam Kidron
[1]Tianjin Institute of Pharmaceutical Research Co. Ltd, China; [2]Preclinical Services, Israel; [3]Oramed Pharmaceuticals, Israel

BACKGROUND

Due to its direct delivery to the hepatic portal system, orally delivered insulin harbors the potential to better mimic physiologically secreted insulin, when compared to subcutaneously delivered insulin. For this same reason, it is projected to have a more favorable impact on hypoglycemia incidence. However, its gastrointestinal absorption is hampered by numerous endogenous enzymatic and mechanical barriers. To this end, insulin was formulated with the unique POD™, which integrates a broad-spectrum protease inhibitor and absorption enhancer to protect the active ingredient and enhance its absorption across the intestinal epithelium.

OBJECTIVES

To assess the pharmacokinetic profile and pharmacodynamic effect of an oral insulin formulation (ORMD-0801), delivered per os (PO) or directly to the duodenum (DU), and to compare them to subcutaneously (SC) delivered insulin.

DESIGN

Four fasting beagle canines were treated with ORMD-0801 (8 mg insulin), per os or duodenally, and with subcutaneous insulin (5 U) on three independent test sessions. Blood samples were drawn over the ensuing 6 h to determine plasma insulin, glucose and c-peptide concentrations. Exogenous insulin concentrations were extrapolated as previously described (Jacobsen et al. Eur J Clin Pharmacol (2000) 56:399).

TABLE 1. PHARMACOKINETIC METRICS

INSULIN		PO		DU		SC	
Parameter	Unit	**Mean**	SD	**Mean**	SD	**Mean**	SD
$t_{1/2z}$	h	0.649	0.508	0.518	0.187	0.99	0.185
C_{max}	μU /ml	625	360	453	436	101.6	45.7
T_{max}	h	0.75	0.289	0.5	0.601	0.375	0.224
$AUC_{(0\text{-}t)}$	μU /ml*h	414	246	423	526	181	42.6
$AUC_{(0\text{-}\infty)}$	μU /ml*h	426	242	433	530	184	41.5
Relative Bioavailability	%	5.41	2.26	8.31	9.94		

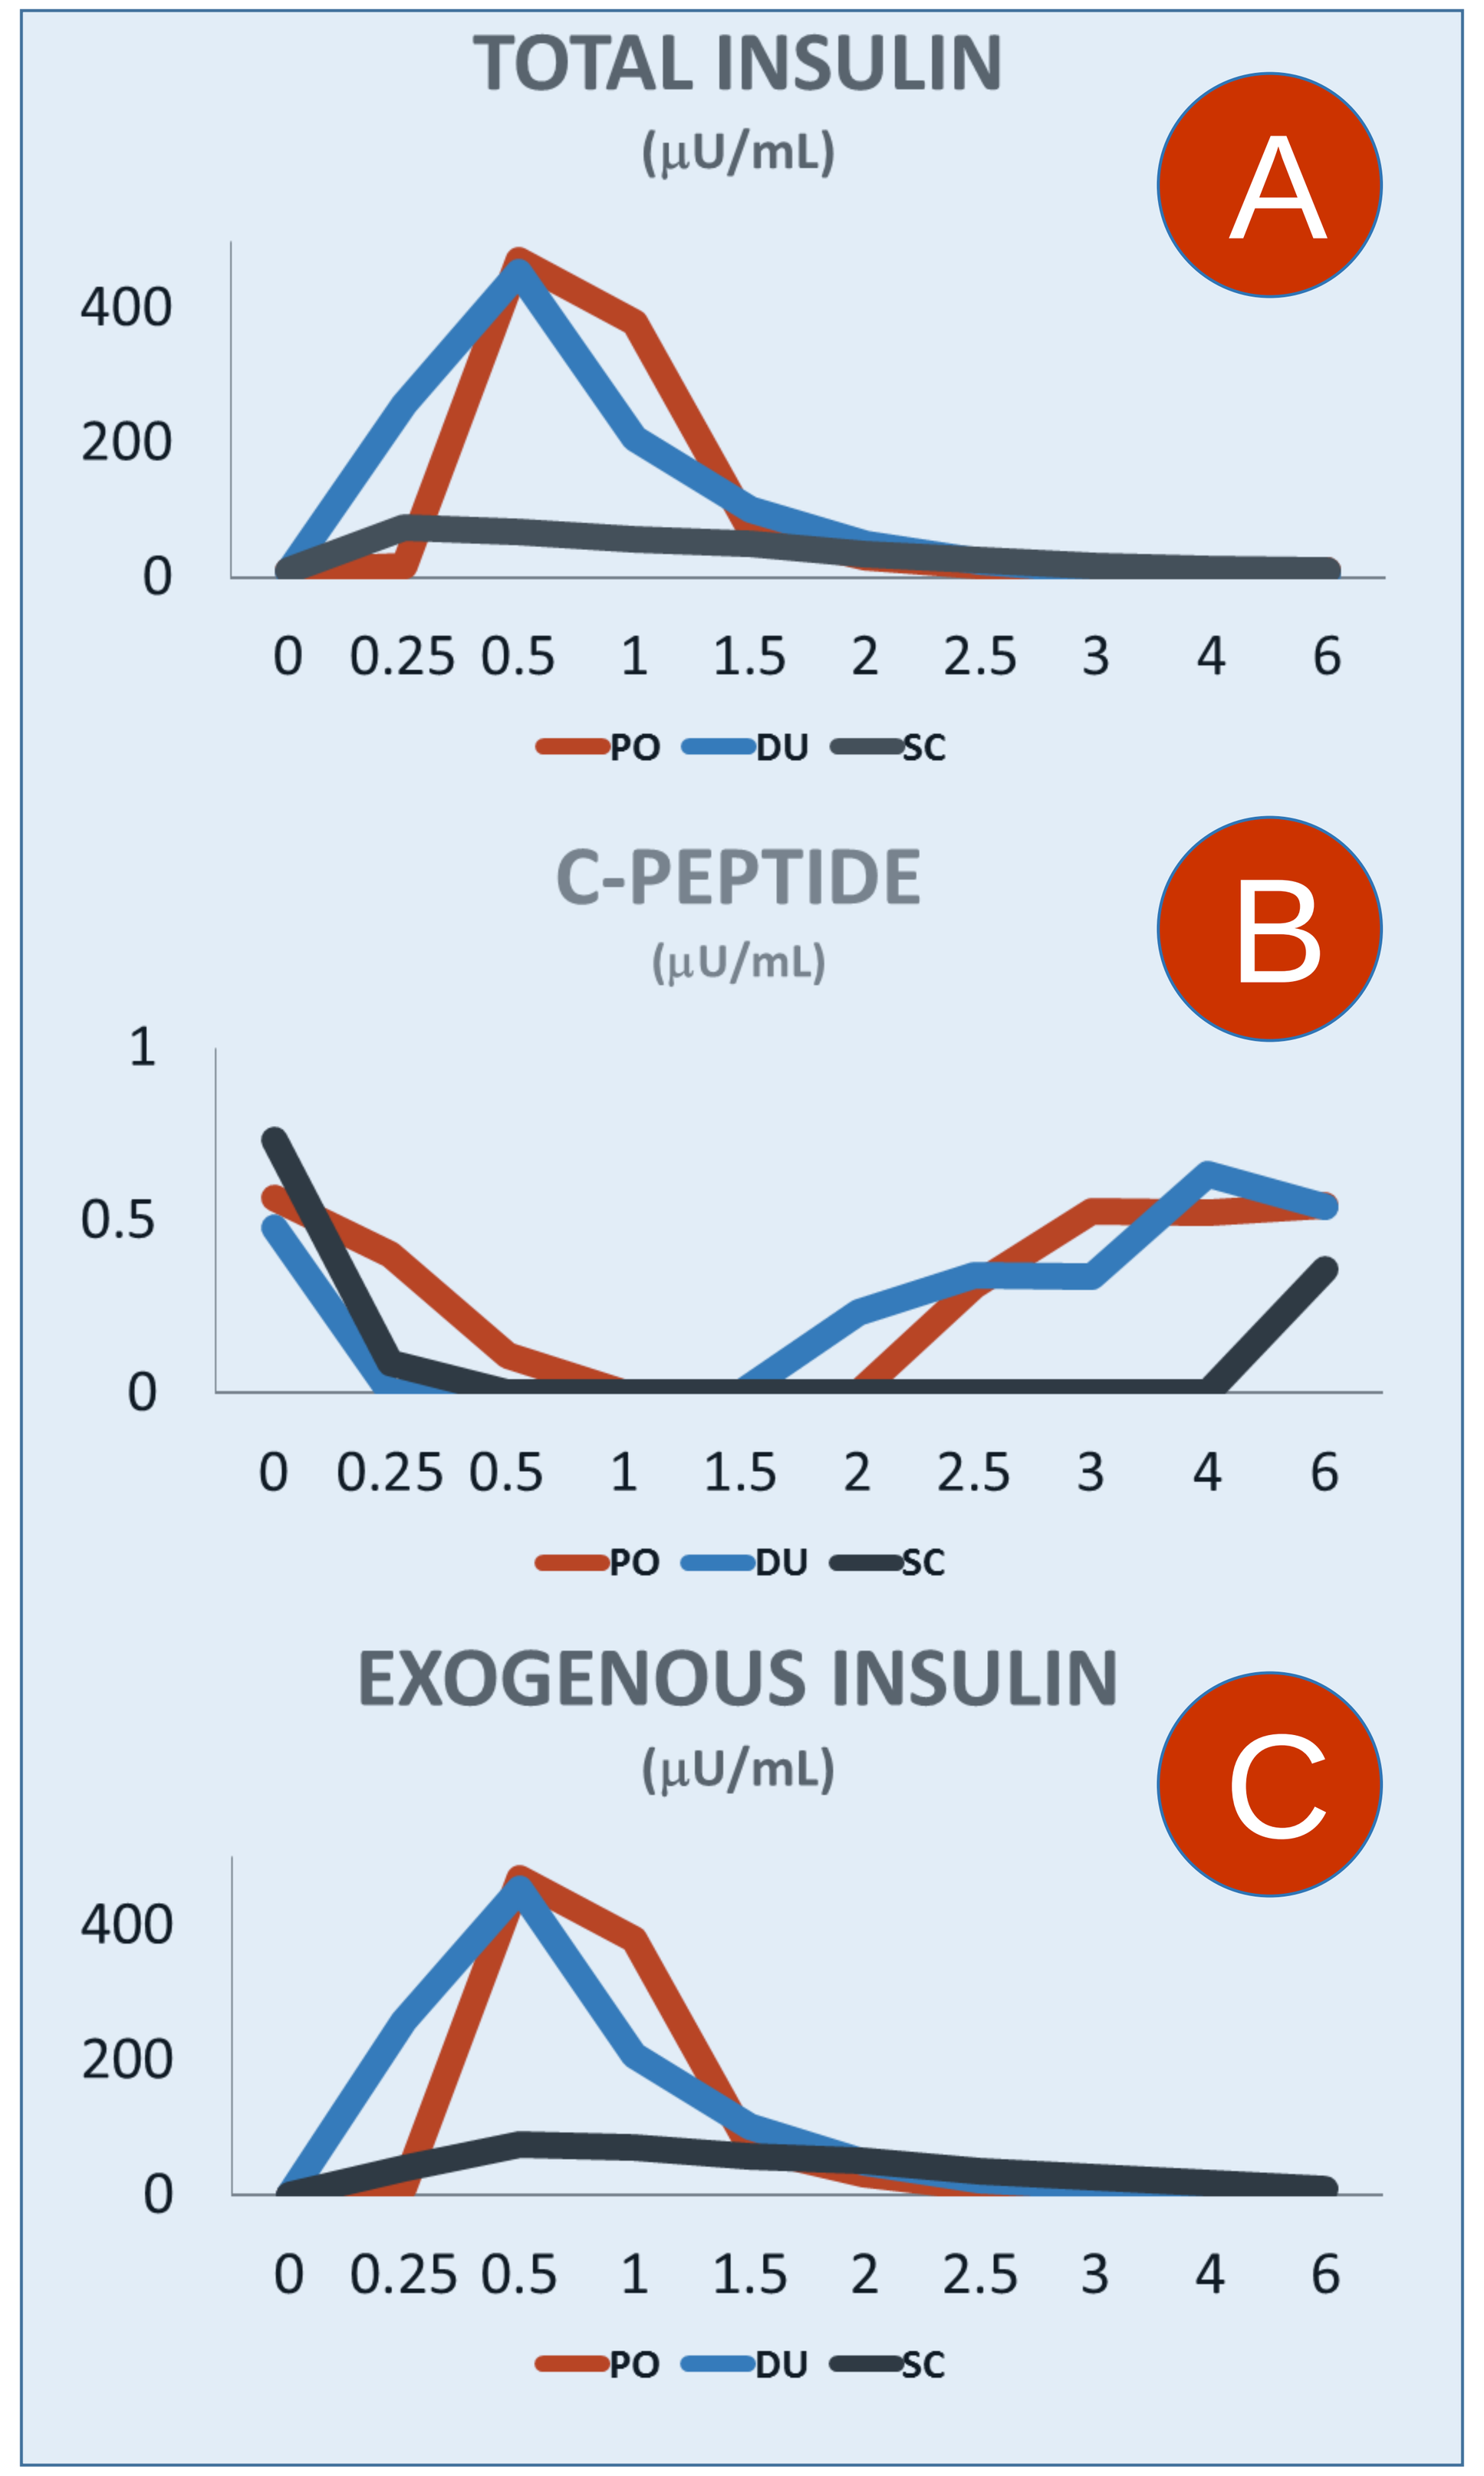
TOTAL INSULIN
(μU/mL)
A
C-PEPTIDE
(μU/mL)
B
EXOGENOUS INSULIN
(μU/mL)
C
PO
DU
SC

Figure 1. Mean insulin profile following oral, duodenal and subcutaneous delivery Four fasting, healthy dogs were treated with ORMD-0801, delivered either orally (PO) or directly to the duodenum (DU), or with subcutaneous (SC) insulin, at three independent test sessions. Blood was periodically collected over the ensuing 6 h period to determine (A) total plasma insulin and (B) c-peptide levels. (C) Exogenous insulin levels were then extrapolated. Mean values at each time point are presented.

1

TABLE 2. PHARMACODYNAMIC METRICS

GLUCOSE		PO		DU		SC	
Parameter	Unit	**Mean**	SD	**Mean**	SD	**Mean**	SD
C_{min}	mg%	25.2	5.4	28.8	10.8	25.2	7.2
T_{min}	h	1.0	0.4	1.5	0.7	1.6	0.9

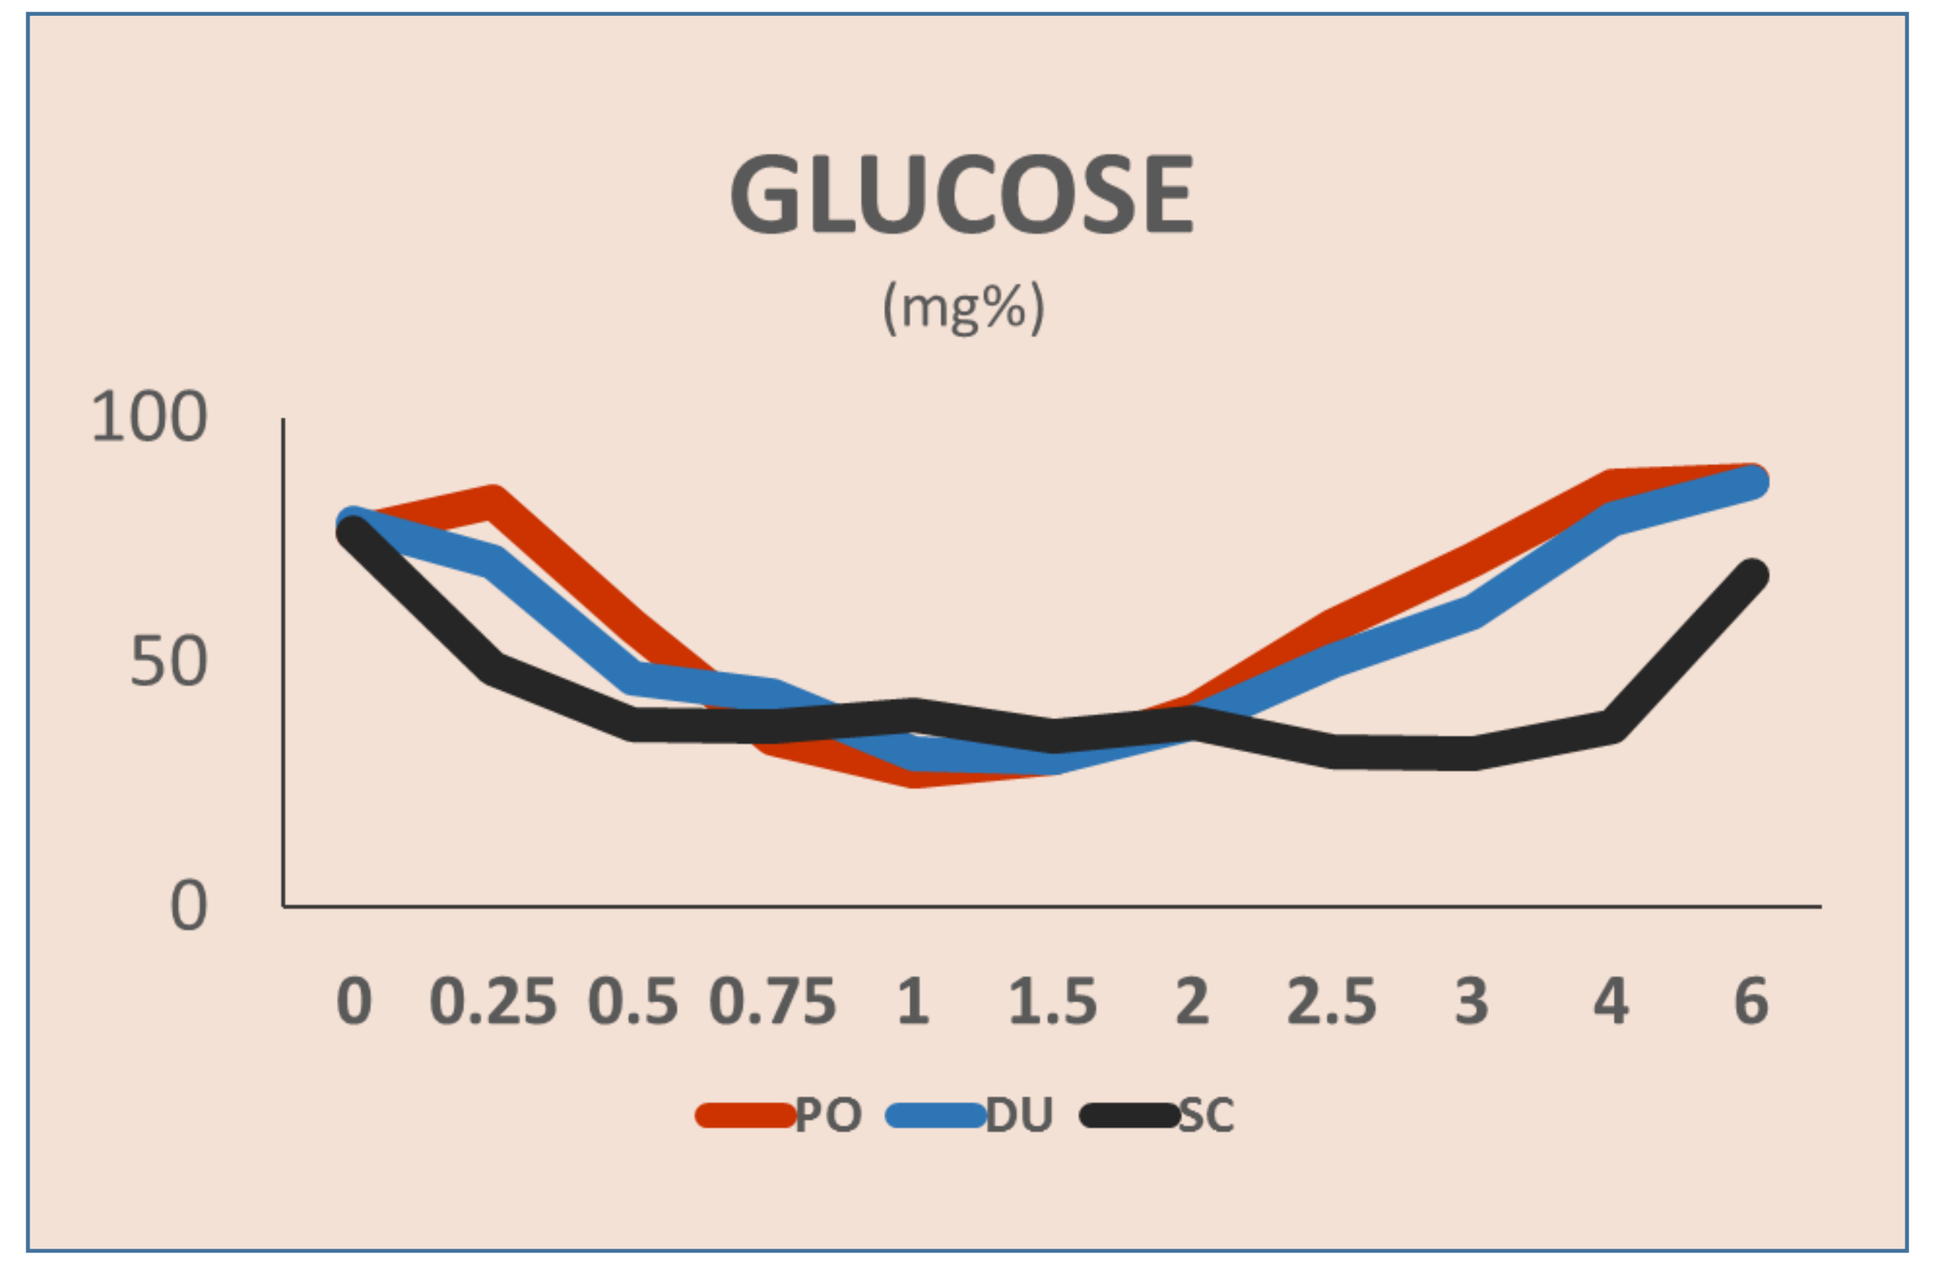
GLUCOSE
(mg%)
PO
DU
SC

Figure 2. Plasma glucose levels following oral, duodenal and subcutaneous delivery Four fasting, healthy dogs were treated with ORMD-0801, delivered either orally (PO) or directly to the duodenum (DU), or with subcutaneous (SC) insulin, at three independent test sessions. Blood was periodically collected over the ensuing 6 h period to determine plasma glucose levels. Mean values at each time point are presented.

2

RESULTS

Maximum exogenous insulin concentrations were highest for PO (625±360 μU/mL), followed by DU (453±436 μU/mL) and SC (101±45.7 μU/mL) insulin (Table 1). Peak absorption was reached within a mean 0.75±0.29h and 0.5±0.6 h in the PO and DU sessions, respectively, and at 0.38±0.22 h for the SC insulin (Table 1). Mean exogenous insulin area under the curve was similar between the PO and DU sessions (414±246 μU/ml*h and 423±526 μU/mL*h, respectively) and significantly higher than after SC delivery (181±42.6 μU/ml*h) (Table 1, Figure 1C). Relative bioavailability of PO insulin and DU insulins was 5.4±2.3% and 8.3±9.9%, respectively (Table 1). Onset of action was typically immediate for the SC insulin and with a lag of 15 min upon PO and DU delivery (Figure 2). Overall the glucose-lowering effect was similar across delivery modes, with minimum levels reached 1.0-1.6 h after dosing (Table 2), but was more prolonged for subcutaneously delivered insulin.

CONCLUSIONS

Oral delivery of insulin with the POD™ technology is feasible and effective, characterized by a rapid onset and short duration of action.

For more information: estee@oramed.com
U.S.:1-646-240-4193; Intl.:+972-2-566-0001

oramed Pharmaceuticals Inc

www.oramed.com