Enteral Administration of Exenatide-4; Proof of Concept Pharmacodynamic Study in Dogs

Miriam Kidron PhD(1, 3), Yael Shushlav DVM(3), Oded Ovadia MSc(2). Ehud Arbit MD(3)

(1)Diabetes Unit, Hadassah University Hospital, (2)School of Pharmacy Hebrew University and (3)Oramed Pharmaceuticals Jerusalem Israel

Diabetes Technology Meeting November 2008

oramed Pharmaceuticals Inc

Introduction:

Exenatide is a synthetic version of exendin-4, a GLP-1 analogues or mimetic and a functional agonist of the GLP-1 receptor. The antihyperglycemic effects of Exenatide are due to its insulinotropic effect as it stimulates glucose-dependent insulin release from the pancreatic islets and its effects of slowing gastric emptying, inhibiting inappropriate glucagon release, stimulating ß-cell proliferation and differentiation, and improving satiety. In clinical trials in patients with T2DM Exenatide when given in combination with metformin and/or sulfonylureas resulted in a hemoglobin A1c (HbA1c) reduction of 1.0% compared with placebo treatment, with the predominant effect on lowering postprandial glucose with less prominent reduction in fasting glucose. Exenatide as well as all other GLP-1 analogues are administered as subcutaneous injections. Exenatide is typically injected twice daily in doses of 5 to 10 µg. A non-parenteral route to administer GLP-1 analogues including Exenatide will have significant therapeutic benefits , being more convenient it will foster compliance and adherence. Oramed is developing an oral dosage form of Exenatide based on its proprietary drug delivery technology, which facilitates the absorption of peptides and proteins across biological membranes. The objective of this study was to establish a dose response to escalating doses of Exenatide in dogs.

Methods:

Study was conducted in 4 beagle dogs with an average weight of 10 kg.. All the dogs had a cannula residing in the jejunum through which the drug was administered. After an overnight fast, the dogs were given different doses of oral GLP-1 analogue or sc injection of the analogue . Absorption of the GLP-1 analogue was assessed by measuring the effect on glucose excursion following an oral glucose load. Control experiment consisted of oral dosing without administration of GLP-1 analogue. The interval between oral administration and the oral glucose load was 30 minutes. The primary efficacy end point was the glucose excursion above the pre-OGTT glucose level over a 150 min interval (incremental area under the curve (AUC) $_{0–150\ min}$.)

Results:

Direct jejunal instillation of GLP-1 analogue significantly (ss) curbed glucose excursion, post glucose load (both in comparison to placebo and among the separate groups)

Table :

OGTT - Glucose AUC$_{0-150\ min}$ Mean ±SD	
Placebo	8906±1508
GLP-1 2.5 µg sc	3656±510
GLP-1 75 µg PO	6292±1043
GLP-1 100 µg PO	5085±931

Discussion:

Oral delivery of proteins and peptide drugs remains a major challenge because of their unique physico-chemical and biologic properties. We have demonstrated in preclinical and clinical studies that our proprietary technology can effectively and reliably transport macromolecules including polypeptides and proteins across biological membranes. Moreover, the native compounds retain their biological activity on reaching the systemic circulation. Clinical studies in healthy volunteers and in people with type 1 and type 2 diabetes have shown that Oramed's oral insulin is absorbed and is effective in lowering blood glucose and decrease c-peptide levels.

In the current study in dogs we have clearly demonstrated that an oral GLP-1 analogue, exenatide, when administered before a meal can blunt meal induced glycemic excursion by about 40% as compared to parenteral exenatide 50% blunting capacity. Pd effects are commonly used in a semi-quantitative way to establish GLP-1 levels in studies assessing DPP IV inhibition. In this study we have demonstrated that the GLP-1 analogue exenatide can be created in an oral dosage form and that it could be ingested by the patient shortly before a meal. These two qualities in a drug significantly facilitate its acceptance among patients and foster higher compliance and adherence to the medication.

Acknowledgments:

Authors would like to thank Tara Horn for her assistance.

Confidential & Proprietary

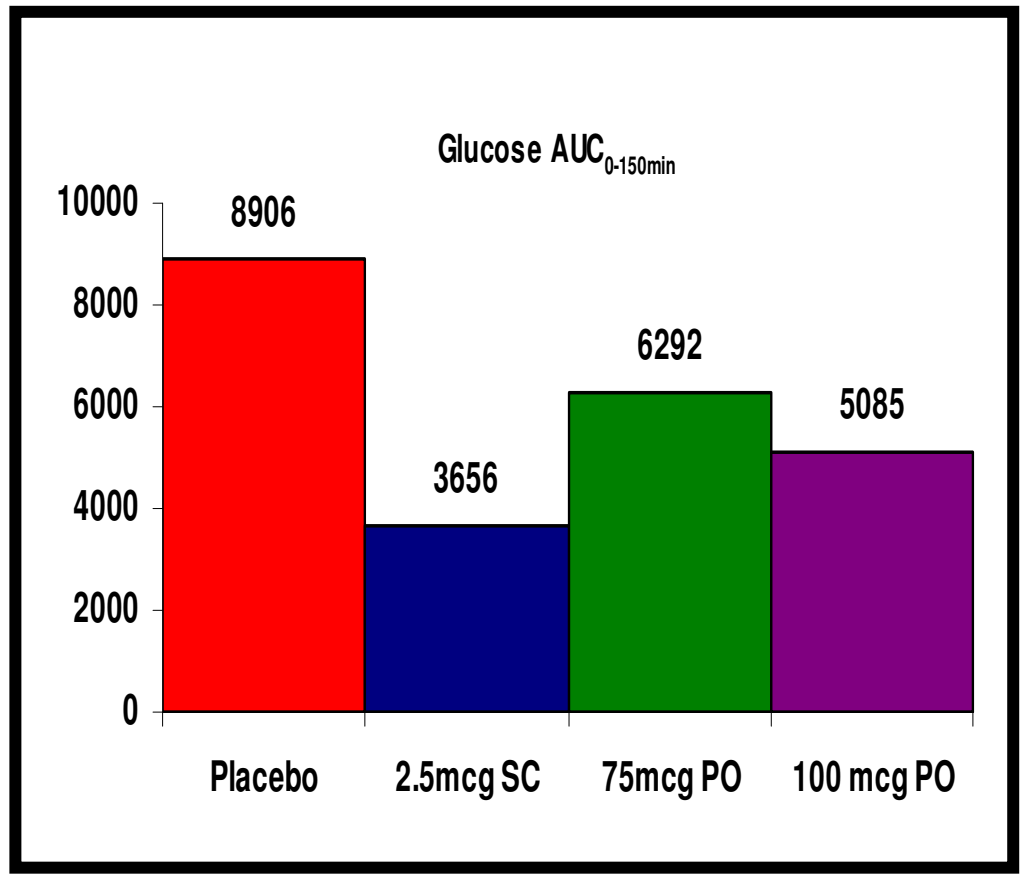
Glucose AUC$_{0-150min}$
10000
8000
6000
4000
2000
0
8906
3656
6292
5085
Placebo
2.5mcg SC
75mcg PO
100 mcg PO

Fig 1 Glucose AUC

Conclusions:

The results of this study in dogs showed that GLP-1 analogue exenatide when combined with Oramed's drug delivery enhancers and formulated in a capsule is absorbed and results in significant blunting of glucose excursion after an oral OGTT. The Pharmacodynamic response to oral exenatide ingestion was robust and reproducible and the short interval between capsule ingestion and meal suggests that a practical and patient friendly oral dosage form can be created. As of now the only incretin mimetics available as oral medication are the DPP IV inhibitors. An oral dosage form of GLP-1 analogues will broaden the choice of available drugs from this important class of antihyperglycemic medication.

For More Information:
Eric Rosenberg
Tel:1-646-240-4193
info@oramed.com
www.oramed.com