Bedtime oral insulin lowers fasting blood glucose levels in T2DM patients

[1]Joel Neutel, M.D.; [2]Miriam Kidron, Ph.D.; [2]Ehud Arbit, M.D.; [3]Kenneth Homer, M.S.
[1] St. Joseph Hospital, Tustin, CA, USA; [2]Oramed Pharmaceuticals, Jerusalem, Israel; [3]Integrium, LLC, Cedar Knolls, NJ, USA

74th scientific sessions
JUNE 13-17, 2014 • SAN FRANCISCO, CA

BACKGROUND

Bedtime insulin administration has been suggested to best counteract abnormal morning fasting blood glucose (FBG) levels, a harbinger of diabetes and a key obstacle to optimal glycemic management in T2DM patients. However, many early-stage patients resist introduction of insulin injections into their routine. The pursuit of an orally bioavailable insulin formulation has been driven by the notion that it can both increase patient compliance, and better mimic the physiological route of naturally secreted insulin, consequently lowering risk of systemic hyperinsulinemia and consequential hypoglycemia. Due to its delayed onset of action, ORMD-0801, an oral insulin formulation developed by Oramed Pharmaceuticals, is well-suited for the control of fasting blood glucose.

OBJECTIVES

To assess the safety and tolerability of bedtime ORMD-0801 in T2DM patients.
To characterize the pharmacokinetics (PK) of bedtime ORMD-0801
To evaluate the pharmacodynamic (PD) effects of bedtime ORMD-0801 on c-peptide levels and on morning fasting glucose levels, compared to placebo

DESIGN

In this randomized, double-blind, placebo-controlled study, 30 adult T2DM patients inadequately controlled with diet and exercise and/or metformin, were outfitted with a blinded continuous glucose monitor (CGM), after a 5-day placebo, outpatient run-in period. On day one, a single placebo dose was administered at bedtime, in an inpatient setting. During the subsequent 7-day period, patients (n=10/cohort) were treated with either 460 IU or 690 IU insulin, or placebo at bedtime. Plasma insulin and c-peptide levels were monitored for 5 hrs thereafter and their values on Day 1 (placebo) were compared to those of Day 8 (treatment or placebo).

1

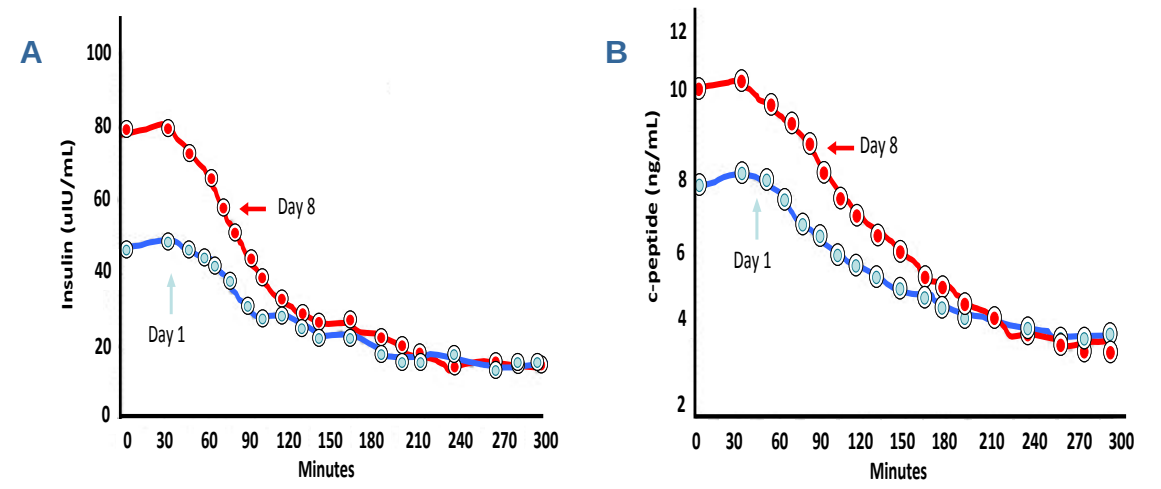
A
Insulin (uIU/mL)
Day 8
Day 1
Minutes
B
c-peptide (ng/mL)
Day 8
Day 1
Minutes

Figure 1. Day 8 vs. Day 1 mean insulin and c-peptide levels. Blood was drawn at set intervals during the five-hour postdosing period. Insulin (A) and c-peptide (B) levels measured on Day 1 (placebo treatment) and Day 8 (following 7 days of ORMD-0801) treatment) are presented.

2

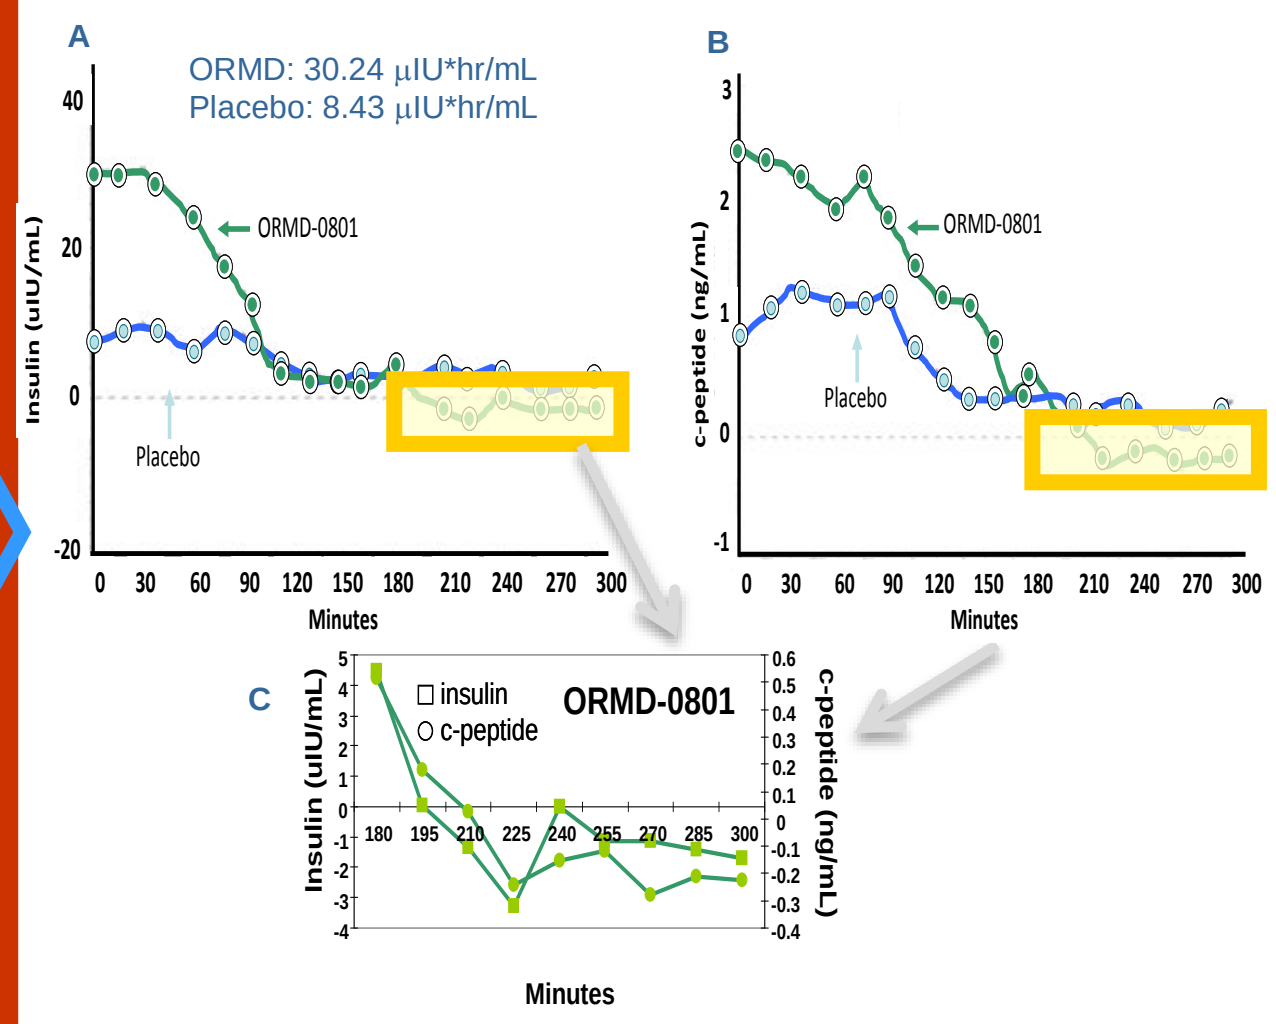
AUC 0-60
A
ORMD: 30.24 μIU*hr/mL
Placebo: 8.43 μIU*hr/mL
Insulin (uIU/mL)
ORMD-0801
Placebo
Minutes
B
c-peptide (ng/mL)
ORMD-0801
Placebo
Minutes
C
insulin
c-peptide
ORMD-0801
Insulin (uIU/mL)
c-peptide (ng/mL)
Minutes

Figure 2. Mean insulin and c-peptide levels in ORMD-0801- vs. placebo-treated T2DM patients. The mean Day 8 vs. Day 1 differences in insulin (A) and c-peptide (B) levels measured over the five-hour postdosing period, were calculated and compared between the placebo and ORMD-0801 arms of the study (n=10 per cohort). (C) Magnification of the response to ORMD-0801 treatment between 180-300 min postdosing.

Table 1. Mean nighttime, daytime and fasting glucose concentrations

Nighttime CGM Glucose (mg/dL)	Placebo N=10 Mean (SD)	ORMD-0801 N=10 Mean (SD)	Daytime CGM Glucose (mg/dL)	Placebo N=10 Mean (SD)	ORMD-0801 N=10 Mean (SD)	Fasting CGM Glucose (mg/dL)	Placebo N=10 Mean (SD)	ORMD-0801 N=10 Mean (SD)
Last 2 days	167.95 (64.17)	135.64 (39.40)	Last 2 days	176.06 (63.70)	153.23 (40.16)	Last 2 days	156.26 (58.62)	126.02 (27.26)
All 7 days	165.85 (60.76)	139.73 (38.86)	All 7 days	175.99 (61.12)	152.55 (23.44)	All 7 days	154.37 (57.99)	129.27 (27.43)

RESULTS

A manufacturing fault limited the efficacy of the 690 IU dose; the data were excluded from the analysis. No hypoglycemic events or any severe or serious adverse events were recorded throughout the entire study period. ORMD-0801-treated patients showed consistently higher mean plasma insulin (Figure 1A) and c-peptide levels (Figure 1B) throughout the 180 min Day 8 postdosing period, when compared to baseline (Day 1). Moreover, in the first 60 min postdosing, plasma insulin exposure was 20.53 mIU*h/mL greater among ORMD-0801-treated patients when compared to the placebo arm (Figure 2A) and followed a concentration-time course similar to that of plasma c-peptide (Figure 2B). In the second half of the monitoring period, c-peptide levels continued to decline, despite steady insulin levels, suggesting absorption of the exogenous insulin and a consequential decline in endogenous insulin levels (Figure 2C). Fasting (5AM-7AM) CGM data demonstrated a mean -30.24 mg/dL difference between the last two days of active versus placebo treatment (Table 1). Similarly, mean nighttime (10PM-6AM) and daytime (6AM-10PM) glucose levels were consistently lower among ORMD-0801-treated vs. placebo-treated patients (Table 1).

CONCLUSIONS

Overall, ORMD-0801 led to a stable, consistent and a short-lived rise in plasma insulin levels, which positively impacted FBG concentrations in the treated T2DM patients. Hepatic insulin resistance, resulting in excessive hepatic glucose production and subsequent fasting hyperglycemia, can be mitigated by small quantities of insulin delivered directly to the liver. Portal delivery of ORMD-0801 is proposed to underlie the observed effect of bedtime dosing on FBG levels.

Note: This trial was a substudy requested by the FDA prior to commencement of a large scale study following a siimilar design. The study was initiated to ensure safety of ORMD-0801 and was not powered to demonstrate efficacy.

For more information: aviva@oramed.com
U.S.:1-718-831-2512; Intl.:+972-2-566-0001

www.oramed.com

oramed Pharmaceuticals Inc