Improved oral insulin bioavailability when delivered in soft capsules

[1]Miriam Kidron, Ph.D.; [2]Camil Fuchs, Ph.D.; [1]Ehud Arbit, M.D.; [3]Shoshi Shpitzen, [3]Daniel Schurr, M.D.
[1]Oramed Pharmaceuticals, Jerusalem, Israel; [2]Department of Statistics, Tel Aviv University, Tel Aviv, Israel; [3]Hadassah Medical Center, Diabetes Unit, Jerusalem, Israel

74th scientific sessions
JUNE 13-17, 2014 · SAN FRANCISCO, CA

BACKGROUND

One of the established clinical advantages of soft gel capsules is the potential to enhance active ingredient bioabsorption and bioavailability, which often translates to lower required drug doses. In addition, they maintain dose uniformity for low-dose drugs and are simpler to swallow, when compared to hard gelatin capsules.

OBJECTIVE

To compare the bioavailability of oral insulin (ORMD-0801) delivered in hard versus soft gelatin capsules.

DESIGN

Following informed, signed consent, one or two hard or soft gelatin enteric-coated capsules containing 8 mg insulin/capsule, were administered to five type 1 diabetes mellitus (T1DM) patients 45-60 min before a standard meal. Plasma insulin concentrations were monitored over the ensuing 5-hour period. The ratios of the mean baseline (0-20 min postdosing) versus treatment (20-300 min postdosing) period insulin levels were calculated.

Table 1. Mean insulin concentration treatment:baseline ratios and AUCs

Insulin	8 mg Soft	8+8 mg Soft	8 mg Hard	8+8 mg Hard
Mean ratio	4.59	5.23	3.51	3.77
Mean AUC (μIU/ml *min^{-1})	1359	1556	1044	1122

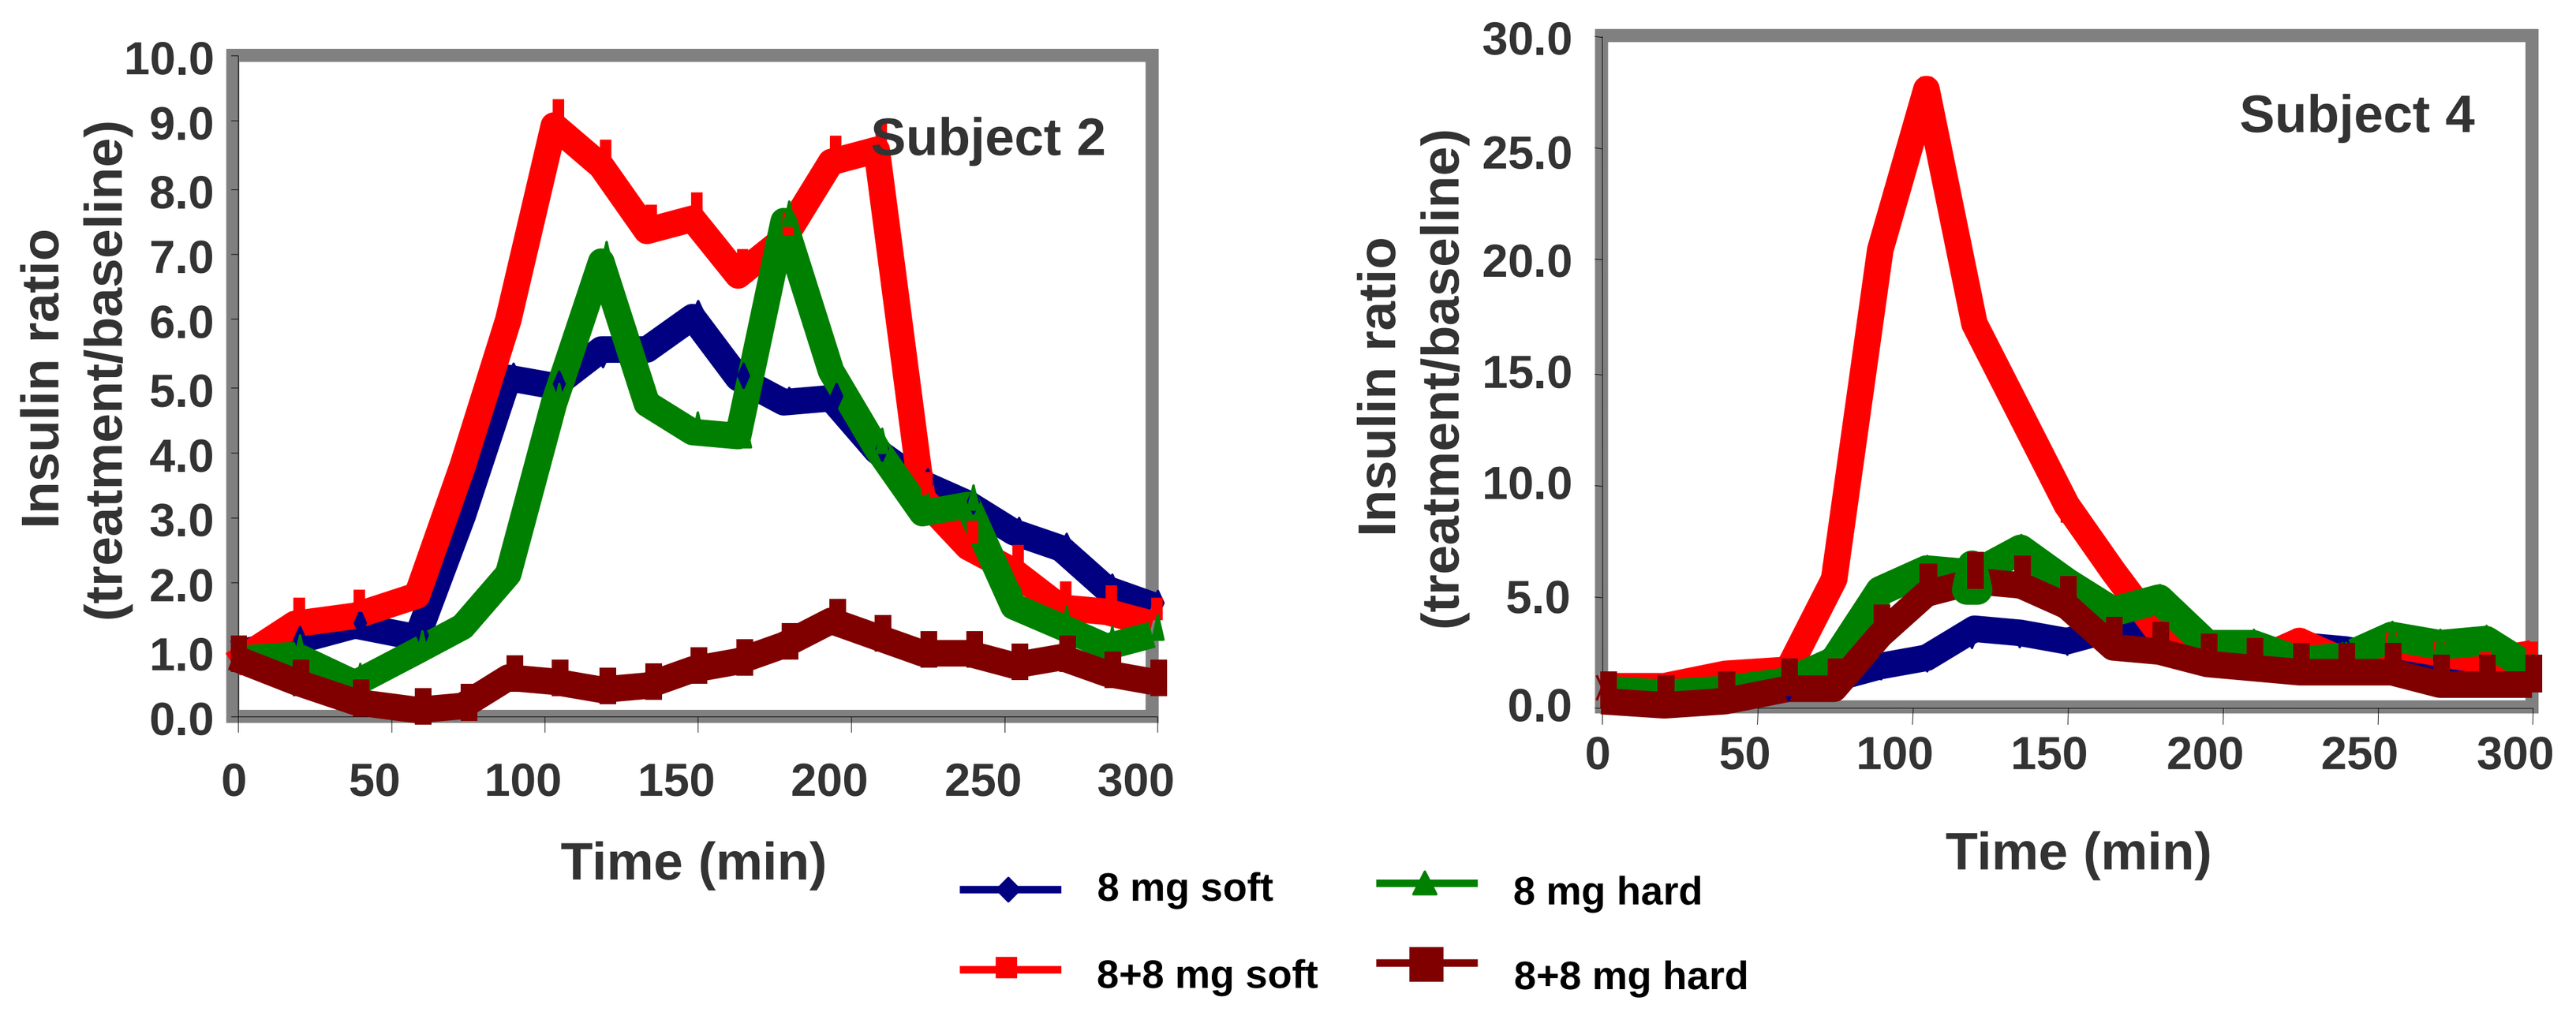
Subject 2
Insulin ratio (treatment/baseline)
10.0
9.0
8.0
7.0
6.0
5.0
4.0
3.0
2.0
1.0
0.0
0
50
100
150
200
250
300
Time (min)
Subject 4
Insulin ratio (treatment/baseline)
30.0
25.0
20.0
15.0
10.0
5.0
0.0
0
50
100
150
200
250
300
Time (min)
8 mg soft
8 mg hard
8+8 mg soft
8+8 mg hard

Figure 1. Insulin concentration treatment:baseline ratios T1DM patients received a dose of 8 or 8+8 mg insulin delivered in soft of hard capsules, 45-60 min before a standard meal. Blood samples were drawn regularly over the five hours thereafter. Treatment:baseline ratios of insulin concentrations were calculated for each blood draw time point.

RESULTS

Higher concentration-time insulin curves were obtained following drug delivery in soft gelatin capsules, when compared to the hard gelatin capsules. The 8 mg dose delivered in a soft capsules was associated with 30.8% and 30.2% higher mean plasma insulin concentrations and area under the curve (AUC) treatment vs. baseline ratios, respectively, when compared to identical doses delivered in hard capsules (Figure 1 and Table 1). Similarly, mean plasma insulin concentration and AUC ratios measured following administration of 8+8 mg insulin in soft gel capsules were 38.6% and 38.9% higher, respectively, than when delivered in a hard gel capsule. Upon dose doubling, mean plasma insulin concentrations and AUC ratios increased by 13.8% and 14.5%, respectively, when delivered in soft capsules, but only by 7.4% when delivered in hard gelatin capsules. Surprisingly, mean glucose levels did not follow the same pattern and were lowest following treatment with 8+8 mg insulin delivered in hard capsules.

CONCLUSIONS

Soft gelatin capsules enhanced the bioavailability of orally delivered insulin (ORDM-0801) when compared to hard gelatin capsules. These findings will be instrumental in advancing the development of oral insulin.

oramed Pharmaceuticals Inc
www.oramed.com
For more information: aviva@oramed.com
U.S.:1-718-831-2512; Intl.:+972-2-566-0001